F-842
ICAAC '04

In-Vitro Activity of a New Triazole BAL4815, the Active Component of BAL8557 (the Water-Soluble Prodrug) against *Aspergillus* spp.

Authors: P. Warn, A. Sharp, D.W. Denning
University of Manchester, Manchester, UK

Correspondence address of the main author:
P. Warn
University of Manchester
Room 1.800
Stopford Building
Oxford Road
Manchester M13 9PT U.K.
E-mail: Peter.Warn@man.ac.uk

Abstract

Background: BAL4815 is the active component of the antifungal agent BAL8557 (the water-soluble prodrug) of the triazole class with broad-spectrum antifungal activity. We compared the *in vitro* activity of BAL4815 with that of itraconazole (ITC), voriconazole (VOR), caspofungin (CAS) and amphotericin B (AMB) against 118 isolates of *Aspergillus* comprising of four different species *(fumigatus, terreus, flavus niger*); the isolates were pre-selected to include 16 isolates demonstrating resistance to either ITC or AMB.

Methods: Susceptibilities were determined for BAL4815, AMB, ITC and VOR using the microdilution plate modification of the NCCLS M38-A method using RPMI 1640 buffered to pH 7.0 with MOPS, for CAS the method was modified using incubation in a gas mixture of 1% O2, 5% CO2 94% N2 to aid reading. The MIC was taken as the lowest drug concentration that inhibited all growth at 48 hr for BAL4815, ITC, VOR and AMB; for CAS the MIC was taken as the lowest drug concentration that which reduced growth by 80% compared with the drug-free control at 48 hours for BAL4815, ITC, VOR AND CAS; for AMB the MIC was taken as that which inhibited all growth. MFCs (≥99% kill) were also determined for all drugs other than CAS.

Results: For all isolates, geometric mean (GM) MIC values and ranges (in mg/L) were: BAL4815 0.345 and 0.125-2.0, ITC 0.178 and 0.063->8.0, VOR 0.223 and 0.125-4.0, CAS 0.341 and 0.125-4.0, AMB 0.450 and 0.06-4.0. No significant differences in susceptibility to BAL4815 were seen between species and in contrast to ITC no isolates demonstrated MICs greater than 2.0mg/L. For all isolates, GM MFC values and ranges (in mg/L) were: BAL4815 1.68 and 0.25->8.0, ITC 1.78 and 0.06->8.0, VOR 1.09 and 0.25->8.0, AMB 0.98 and 0.25->4.0. A reproducibility study on 20% of the isolates showed that 87.5%, 87.5%, 87.5%, 91.7% and 95.8% of isolates retested were within one well of the original MIC value for BAL4815, ITC, VOR, CAS and AMB, respectively.

Conclusions: BAL4815 demonstrated promising antifungal activity against all four *Aspergillus* species *in vitro* including strains resistant to ITC, CAS and AMB and warrants further *in vivo* investigation.

Introduction

Despite advances in antifungal therapy mortality rates following invasive aspergillosis remain unacceptably high.[1]. The new formulations of amphotericin whilst less toxic still have significant side-effects. There are concerns about the use of the echinocandins as first-line therapy in aspergillosis. The characteristics of the azoles now available are less than ideal in terms of efficacy, pharmacokinetic/ pharmacodynamic characteristics, drug interactions and protein binding[2].

BAL4815 is the active component of the antifungal agent BAL8557 (the water-soluble prodrug) of the triazole class with broad-spectrum antifungal activity. We compared the in vitro activity of BAL4815 with that of itraconazole (ITC), voriconazole (VOR), caspofungin (CAS) and amphotericin B (AMB) against 118 isolates of *Aspergillus* comprising of four different species (*fumigatus, terreus, flavus* or *niger*); the isolates were pre-selected to include 16 isolates demonstrating resistance to either ITC, AMB or CAS.

Materials and Methods

Organisms
Susceptibility tests were performed on 118 clinical *Aspergillus* isolates; 62 *A. fumigatus* isolates, 20 *A. flavus* isolates , and 18 isolates each of *A. terreus* and *A. niger*. 16 *A. fumigatus* isolates were resistant *in vitro* to ITC.

Antifungal Agents
BAL4815 (Basilea Pharmaceutica, Basel, Switzerland) was provided as a pure powder from the manufacturer. ITC (Janssen Pharmaceuticals, Beerse, Belgium) and AMB (Sigma, Poole, UK) were obtained as a pure compound. VOR and CAS were obtained in vials for intravenous administration (Salford Royal Hospital, Manchester UK). Stock solutions (3200µg/mL) were aliquoted and stored in glass vials, at -20°C until required.

Susceptibility testing
Susceptibility tests were performed using the broth microdilution modified method of NCCLS M38-A using RPMI 1640 medium buffered to pH 7.0 with MOPS[3]. In brief final drug ranges (in mg/L) were BAL4815, VOR and ITC 0.0078-8, CAS and AMB 0.0039-4. Inoculum suspensions were prepared from day 5 to 8 day cultures grown on Sabouraud dextrose agar at 37°C and adjusted using a counting chamber. The final inoculum was between 0.5 x 10^4 and 5 x 10^4 CFU/mL.
BAL4815, ITC, VOR and AMB microdilution plates were incubated in air; CAS microdilution plates were incubated in 1% O_2, 5% CO_2 94% N_2 to aid reading[4]. Readings were made after 48 h of incubation at 37°C

The MIC endpoints for BAL4815, ITC, VOR and AMB were read visually as the lowest drug concentration that prevents any discernible growth. The MIC endpoints for CAS were read visually and taken as that which reduced growth by 80% compared with the drug-free control. MFCs (≥99% kill) were also determined for all drugs other than CAS by subculturing 100µL from all wells without visible growth. 20% of the isolates were retested against each drug to assess the reproducibility.

Results

- For all isolates, geometric mean (GM) MIC values and ranges (in mg/L) were: BAL4815 0.620 and 0.125-2.0, ITC 0.399 and 0.063->8.0, VOR 0.347 and 0.125-8.0, CAS 0.341 and 0.125-4.0, AMB 0.450 and 0.06-4.0.
- For all isolates, GM MFC values and ranges (in mg/L) were: BAL4815 1.68 and 0.25->8.0, ITC 1.78 and 0.125->8.0, VOR 1.09 and 0.25->8.0, AMB 0.98 and 0.25->4.0.
- In contrast to ITC no isolates demonstrated BAL4815 MICs greater than 2.0 mg/L.
- No significant differences in susceptibility to BAL4815 were seen between species.
- Reproducibility was excellent at ≥87.5% for all organism drug combinations.
- BAL4815 had fungicidal activity against all species of *Aspergillus*

Table 1: In vitro *susceptibilities of 118 isolates of* Aspergillus *to AMB, BAL8415, CAS, ITC and VOR*

Species (no if isolates)	Antifungal agent	MIC (mg/mL)			
		GM	Range	50%	90%
A. fumigatus (62)	AMB	0.334	0.06-0.5	0.25	0.5
	BAL8415	**0.578**	**0.125-2.0**	**0.5**	**2.0**
	CAS	0.437	0.25-4.0	0.5	0.5
	ITC	0.676	0.125->8.0	0.25	>8.0
	VOR	0.334	0.125-8.0	0.25	0.5
A. terreus (18)	AMB	0.735	0.25-1.0	1.0	1.0
	BAL8415	**0.463**	**0.25-0.5**	**0.5**	**0.5**
	CAS	0.354	0.125-1.0	0.5	0.5
	ITC	0.151	0.06-0.5	0.125	0.25
	VOR	0.291	0.25-0.5	0.25	0.5
A. flavus (20)	AMB	0.785	0.5-4.0	1.0	1.0
	BAL8415	**0.732**	**0.5-2.0**	**0.5**	**1.0**
	CAS	0.240	0.125-0.25	0.25	0.25
	ITC	0.138	0.06-0.5	0.125	0.5
	VOR	0.420	0.25-1.0	0.5	0.5
A. niger (18)	AMB	0.412	0.25-1.0	0.5	0.5
	BAL8415	**0.890**	**0.25-2.0**	**0.5**	**2.0**
	CAS	0.206	0.125-0.25	0.25	0.25
	ITC	0.606	0.25-4.0	0.5	2.0
	VOR	0.381	0.25-1.0	0.25	1.0
All isolates (118)	AMB	0.452	0.06-4.0	0.5	1.0
	BAL8415	**0.620**	**0.125-2.0**	**0.5**	**2.0**
	CAS	0.341	0.125-4.0	0.25	0.5
	ITC	0.399	0.06->8.0	0.25	>8.0
	VOR	0.347	0.125-8.0	0.25	1.0

This table is the summary of data using no visable growth MIC endpoints for azoles and amphotericin

Figure 1: *Relationship between MIC and MFC against BAL4815, ITC & VOR*

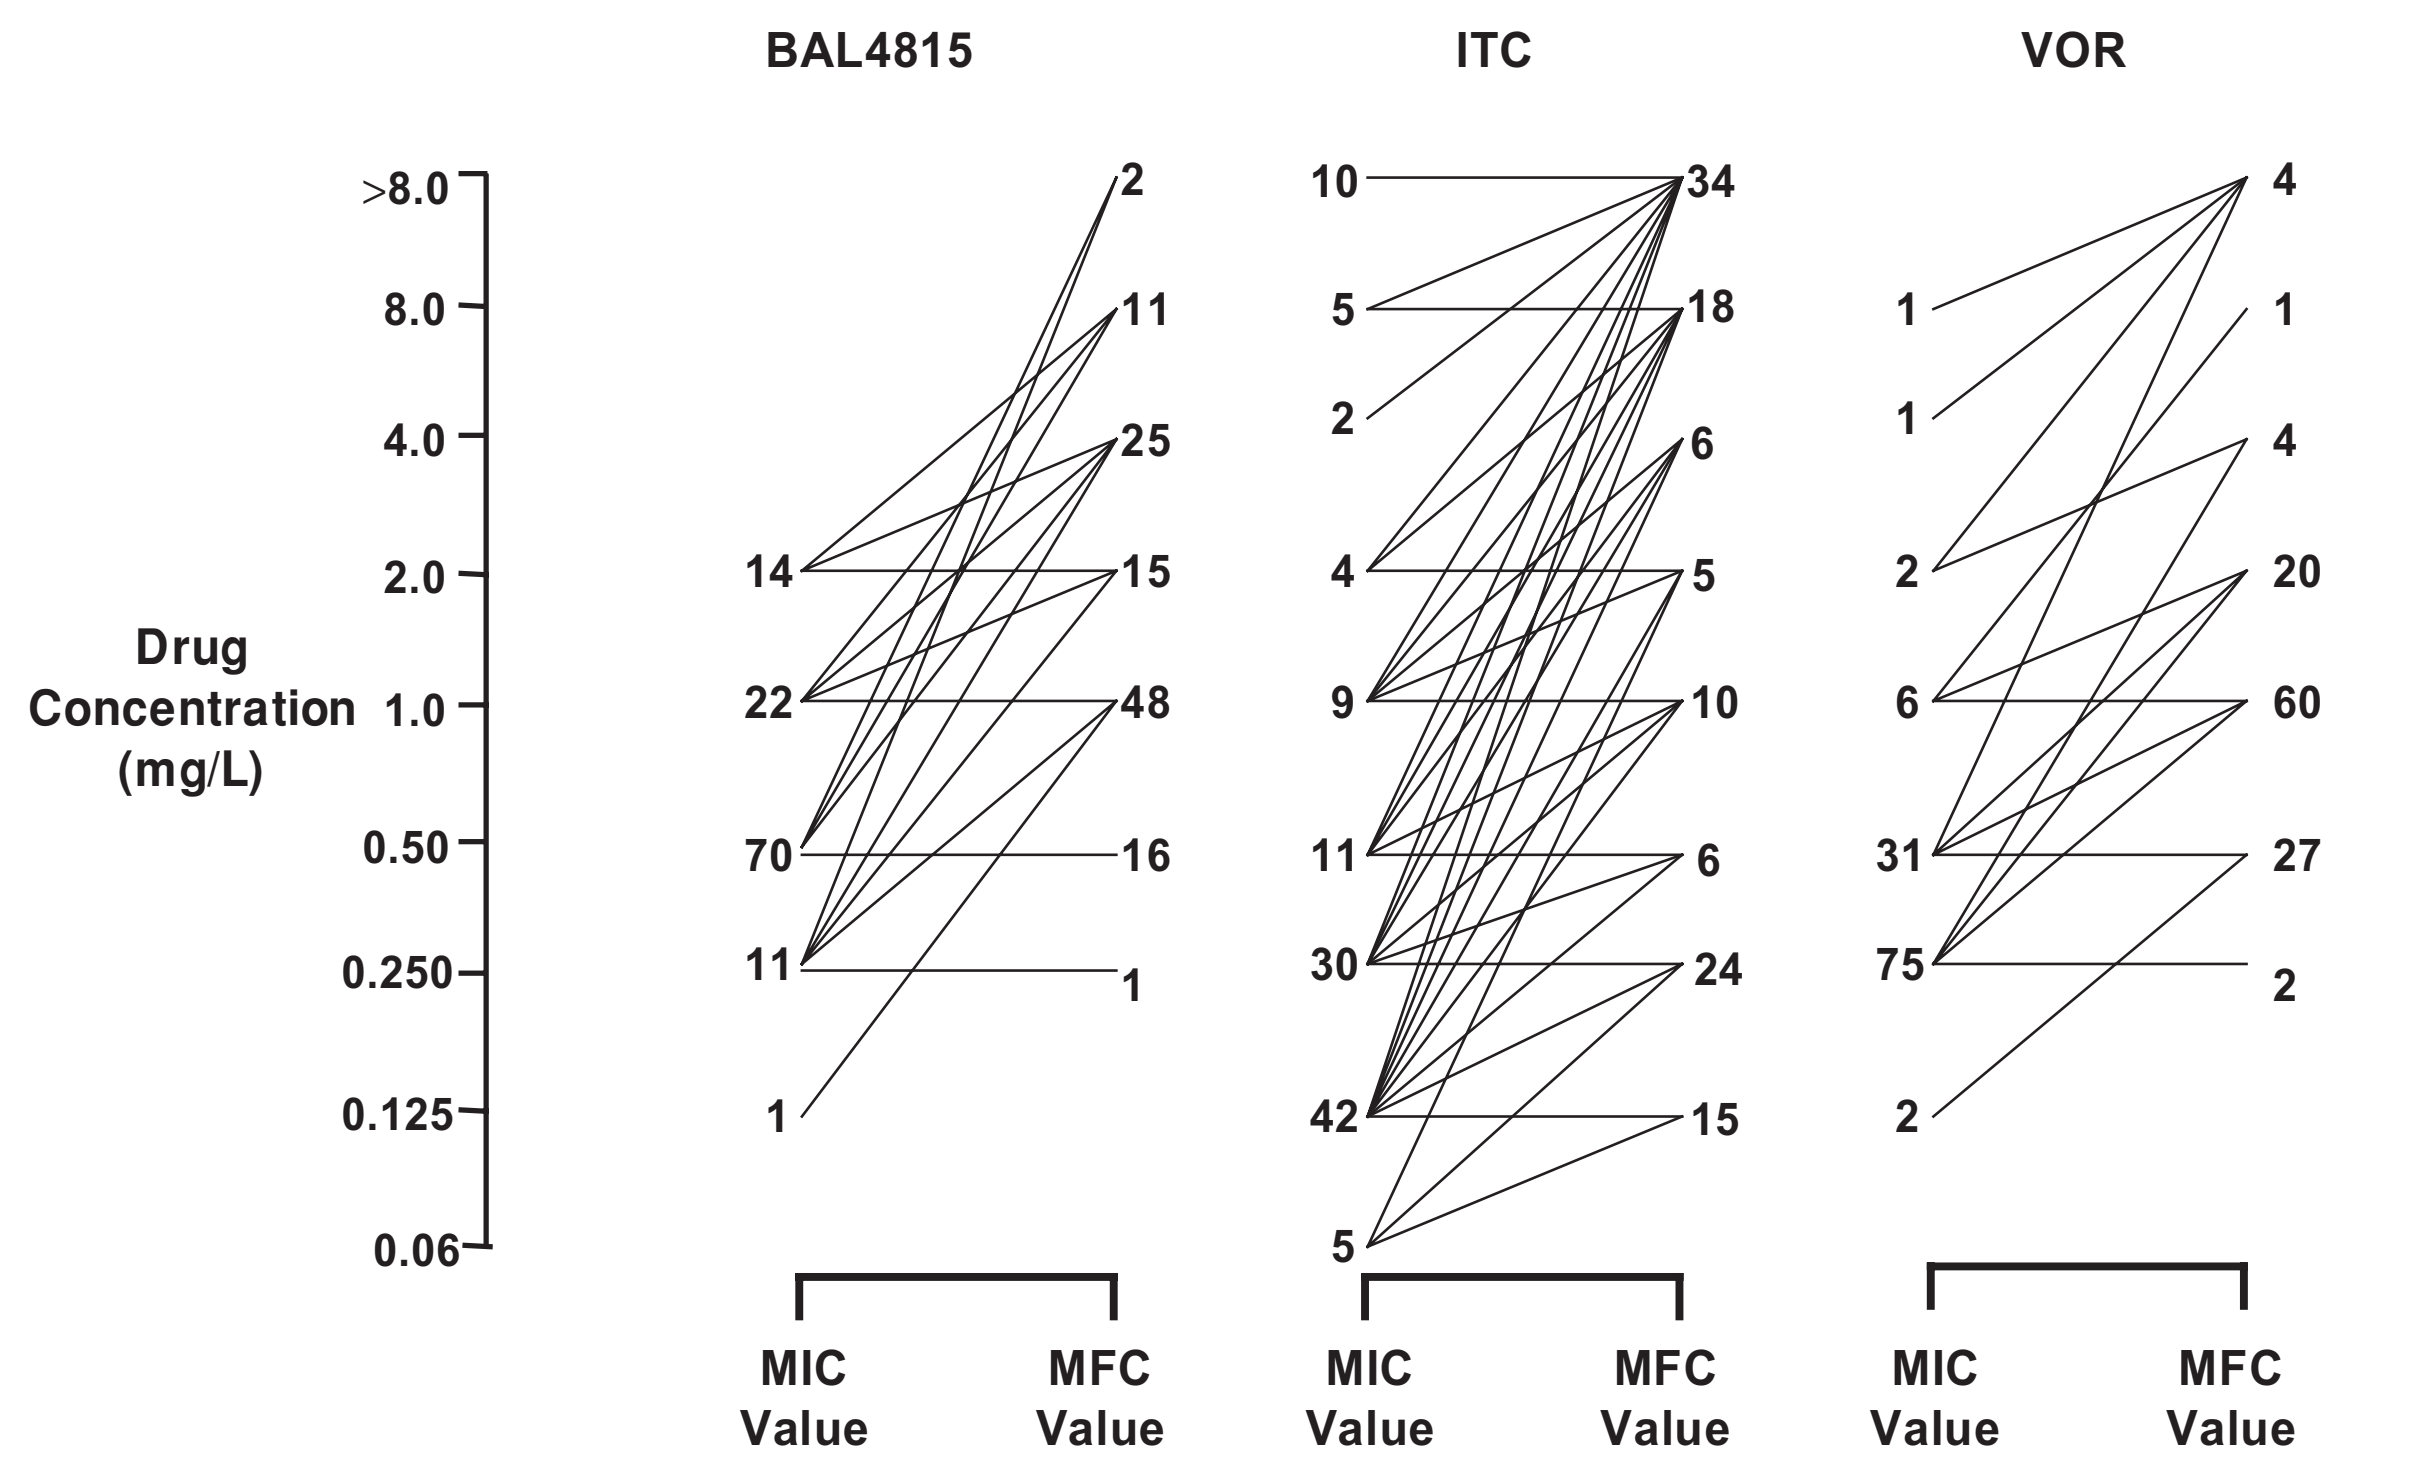

BAL4815
ITC
VOR
Drug Concentration (mg/L)
>8.0
8.0
4.0
2.0
1.0
0.50
0.250
0.125
0.06
14
22
70
11
1
2
11
25
15
48
16
1
10
5
2
4
9
11
30
42
5
34
18
6
5
10
6
24
15
1
1
2
6
31
75
2
4
1
4
20
60
27
2
MIC Value
MFC Value

Conclusions

- *In vitro* susceptibility testing of *Aspergillus* with BAL4815 is possible with reproducible endpoints
- BAL4815 is active against all four *Aspergillus* species *in vitro* including strains resistant to ITC, CAS and AMB.
- No isolates demonstrated MICs to BAL4815 greater than 2mg/L
- BAL4815 shows promising *in vitro* antifungal activity and warrants further *in vivo* investigation

REFERENCES

1. **Herbrecht R, Denning DW, Patterson TF et al.** Voriconazole versus amphotericin B for primary therapy of invasive aspergillosis. New England Journal of Medicine 2002; **347**: 408–415
2. **Hope WW and Denning DW.** Invasive aspergillosis: current and future challenges in diagnosis and therapy. Clinical Microbiology and Infection 2004; **10**: 2–4
3. **National Committee for Clinical Laboratory Standards.** Reference method for broth dilution antifungal susceptibility testing of conidium-forming filamentous fungi. Approved standard. Document M38-A 2002a. NCCLS, Wayne, PA . USA
4. **Warn PA, Sharp A, Guinea J, and Denning DW.** Effect of hypoxic conditions on *in vitro* susceptibility testing of amphotericin B, itraconazole and micafungin against *Aspergillus* and *Candida*. Journal of Antimicrobial Chemotherapy 2004; **53**:743-9

Acknowledgement

P. Warn and A. Sharp are funded by The Fungal Research Trust

This work was part funded by Basilea Pharmaceutica Ltd.